RO-349

Immune monitoring during TOL101- anti-αβ-TCR monoclonal Phase 2 renal transplant clinical trials

Wiseman AC[1], Mulgaonkar S[2], Sung RS[3], Melton LB[4], Waid TH[5], Thompson JS[5], Brown SA[5], Getts MT[6], Frederick TJ[6], Getts DR[6&7] and Flechner SM[8].

[1]University of Colorado Denver, Aurora, CO [2]Saint Barnabas Medical Center, Livingston, NJ;[3] University of Michigan, Ann Arbor, MI; [4]Baylor University Medical Center, Dallas, TX; [5]University of Kentucky, Lexington, KY [6]Research and Development, Tolera Therapeutics, Kalamazoo, MI, [7]Northwestern University, Chicago, IL, [8]Cleveland Clinic Foundation, Cleveland OH.

Introduction

- Current induction agents are far from ideal, and novel agents are needed to overcome numerous issues that occur with the standard of care treatments, including infusion reactions, infections and other safety issues.[1]
- The TCR complex has historically been known as a mitogenic target[1]. However, the αβ T cell receptor is a unique immune target as it lacks immunoreceptor tyrosine activation motifs (ITAMS).
- TOL101 is a specific monoclonal antibody targeting the αβ T cell receptor.
- TOL101 is currently in Phase 2 clinical testing in the USA, evaluating safety and efficacy of TOL101 induction versus anti-thymocyte globulin to prevent kidney transplant rejection
- Primary study objective:-
 1. To assess the safety and tolerability of TOL101 in first time renal transplant recipient
- Secondary study objectives:

1. To assess the pharmacokinetics and the pharmacodynamic effect of TOL101
2. To assess the preliminary clinical efficacy of TOL101
 - Biopsy results, episodes of BPAR, graft and patient survival, DGF, renal function, DSA
3. To investigate the immunogenicity of TOL101

Objectives

- Evaluation of immune parameters both for safety and efficacy
 1. The effect of escalating doses of TOL101 on CD3+T cells (T cell count)
 2. The ability for TOL101 to modulate memory and naïve T cells, as well as other T cell subsets
 3. The evaluation of safety parameters including AE's, cytokine production and HAMA induction

Methods

TOL101 is an investigational drug being tested in the USA under FDA IND#104594

- **Immune monitoring tests**

1.CD3 counts and immuno-phenotyping
2.Cytokine (IL2, IL10,IL1β, IL6,TNF,IFN-γ) analysis
3.HAMA analysis
4.Standard hematological analysis

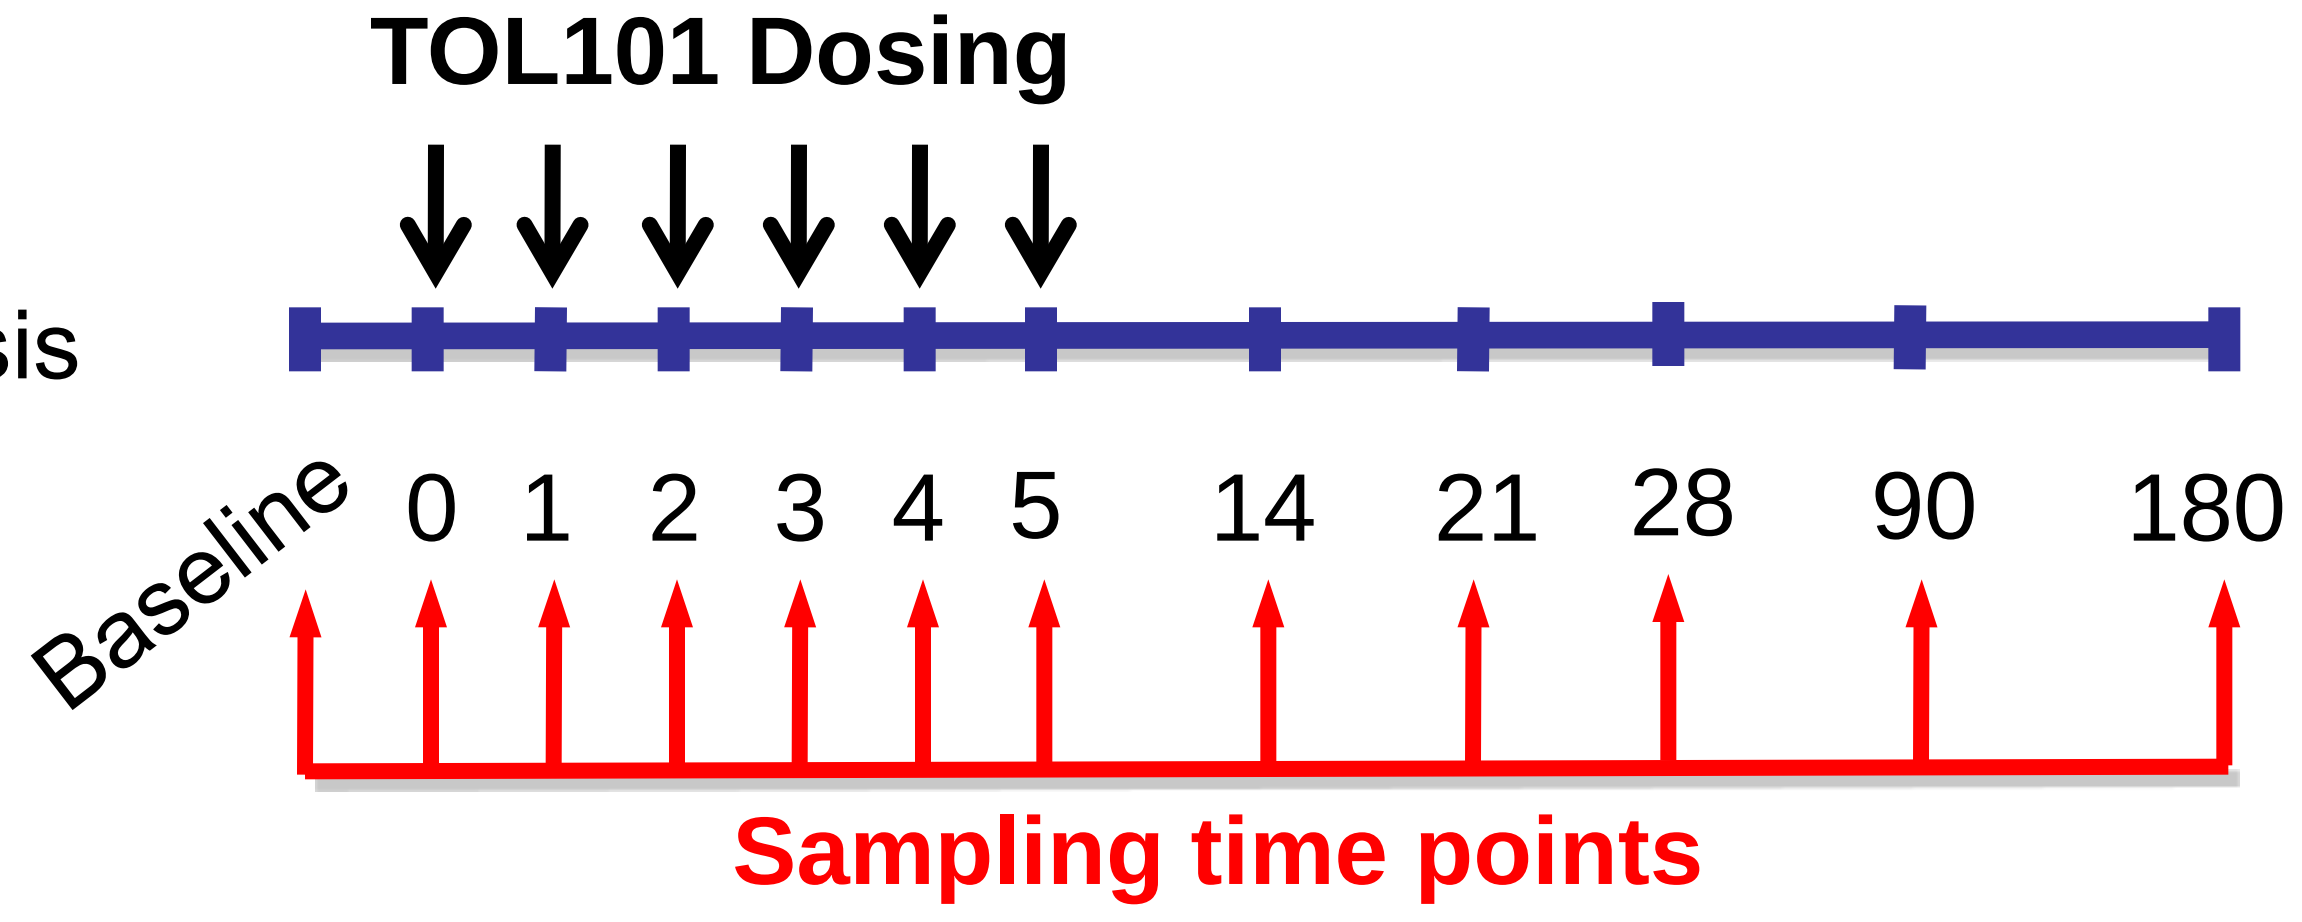

TOL101 Dosing
Baseline
0 1 2 3 4 5 14 21 28 90 180
Sampling time points

- **Brief methods & procedures**

Flow cytometry- T cell counts are obtained from whole blood using the Beckman Coulter Flow-Count Flourospheres procedure. T cell phenotype analysis is conducted on Ficoll separated samples. In the current study, the expression profiles of over 15 proteins are being evaluated; however, here we will discuss the effects of TOL101 on CD3, CD4, CD8, CD45RA and CD45RO. All procedures are performed at a central laboratory.

Multiplexed & Standard ELISA- Multiplexed luminex ELISA is utilized to determine serum cytokine levels. HAMA is determined through standard sandwich ELISA. All procedures are performed at a central laboratory.

Results

- **Dose Dependent CD3 count reduction**

All tested doses resulted in short term reduction
>14mg began to show sustained reduction during dosing

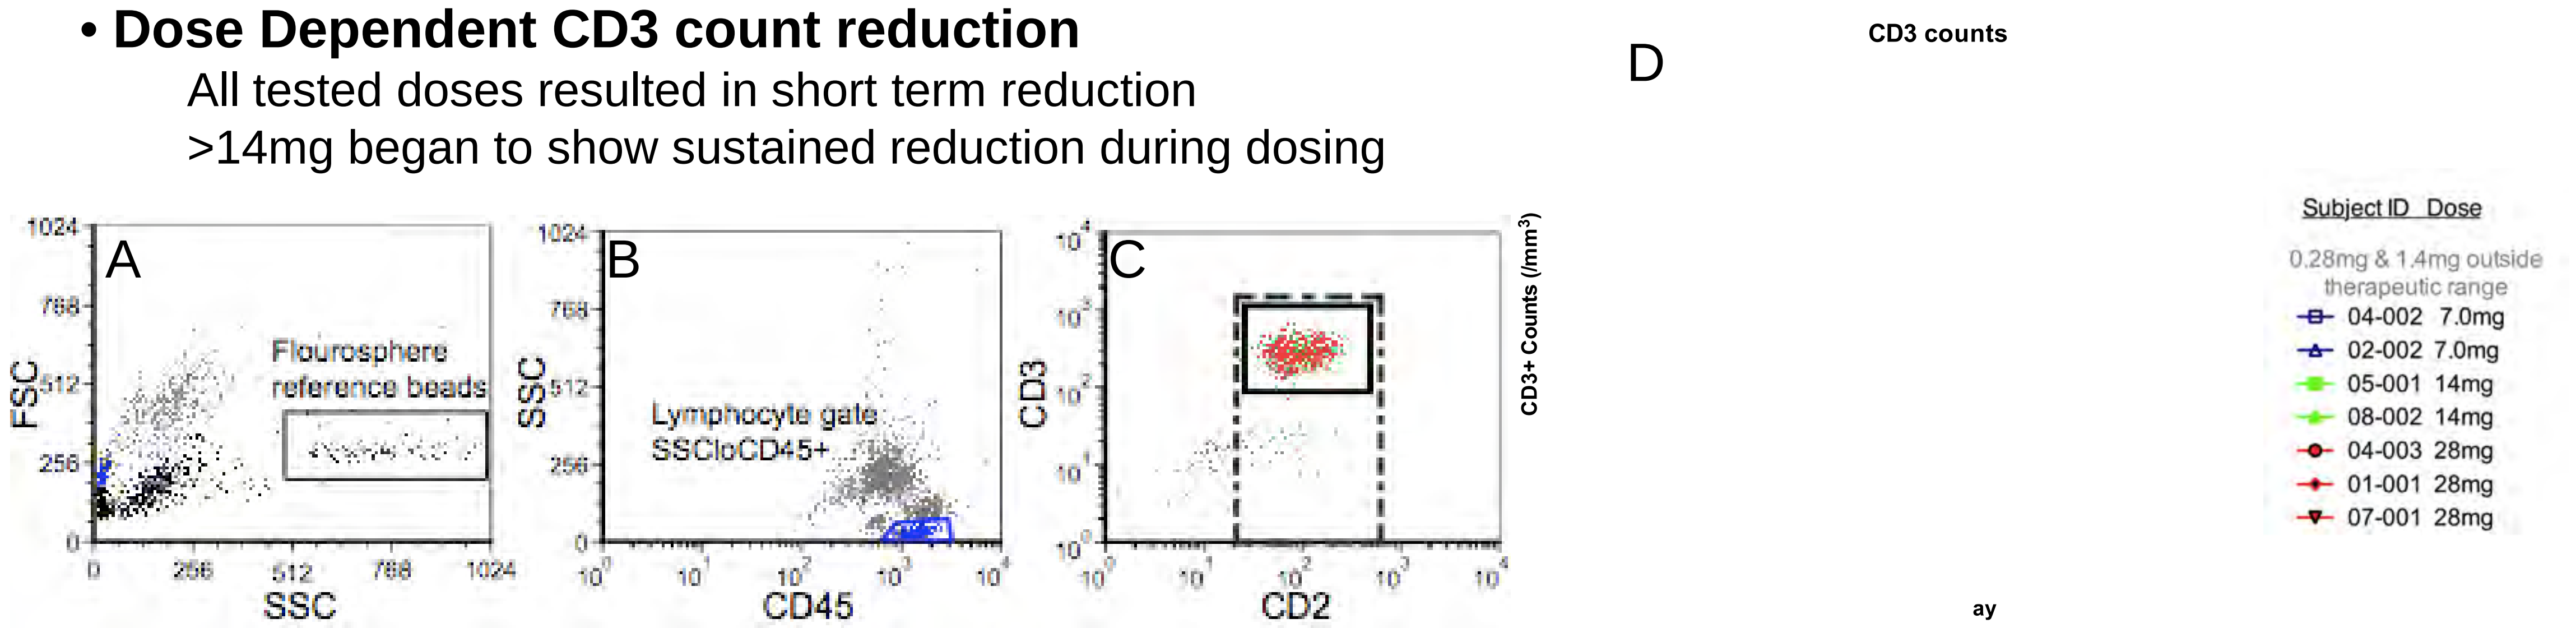

D
CD3 counts
A
FSC
SSC
Flourosphere reference beads
B
SSC
CD45
Lymphocyte gate SSCloCD45+
C
CD3
CD2
CD3+ Counts (/mm3)
Subject ID Dose
0.28mg & 1.4mg outside therapeutic range
04-002 7.0mg
02-002 7.0mg
05-001 14mg
08-002 14mg
04-003 28mg
01-001 28mg
07-001 28mg
ay

Figure 1. Central laboratory flow cytometrical analysis after TOL101 therapy. (A-C), gating strategy for CD3 counts, showing bead standards (A), Lymphocyte gating (B) and CD2 vs CD3 gating (C). Note CD3 counts given by solid box. TOL101 was shown to successfully reduced the CD3 count in a dose-dependent manner.

- **Functional T cell inactivation, not depletion**

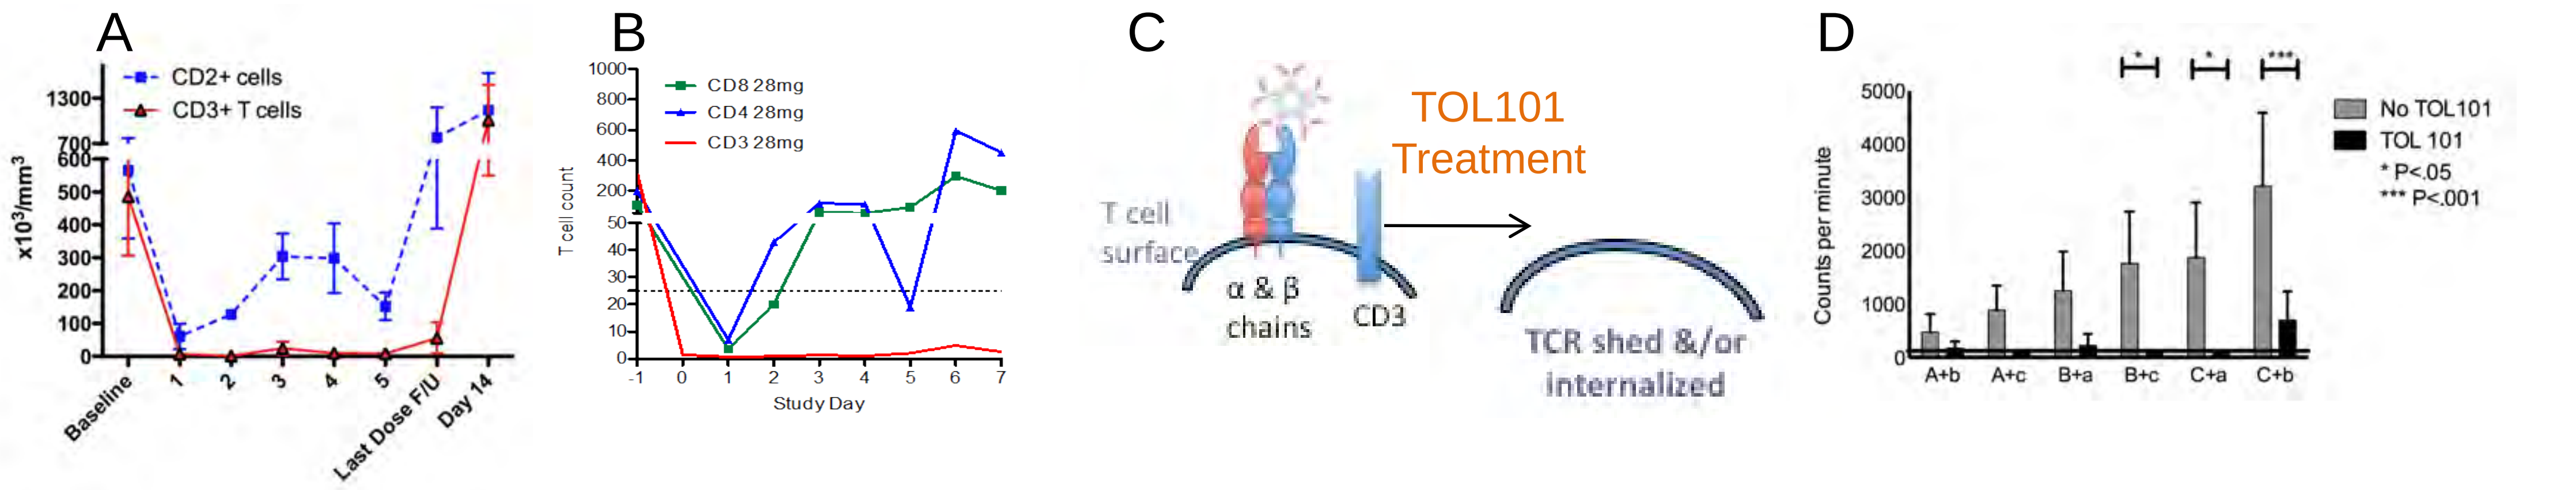

A
CD2+ cells
CD3+ T cells
x10³/mm³
Baseline
Last Dose F/U
Day 14
B
CD8 28mg
CD4 28mg
CD3 28mg
T cell count
Study Day
C
TOL101 Treatment
T cell surface
α & β chains
CD3
TCR shed &/or internalized
D
Counts per minute
No TOL101
TOL 101
* P<.05
*** P<.001
A+b A+c B+a B+c C+a C+b

Figure 2. (A) Examination of CD3+CD2+ T cells and CD3-CD2+ T cells after TOL101 therapy. While the TCR is not expressed CD2+ cells remain. A similar scenario is observed for CD4+ and CD8+ T cells (B). The data support TOL101 removes the TCR complex from the cell surface without depleting the cells (shown schematically in C). This mechanism of action results in strong prevention of the anti-alloantigen response *in-vitro* (D).

- **No cytokine release or HAMA observed to date**

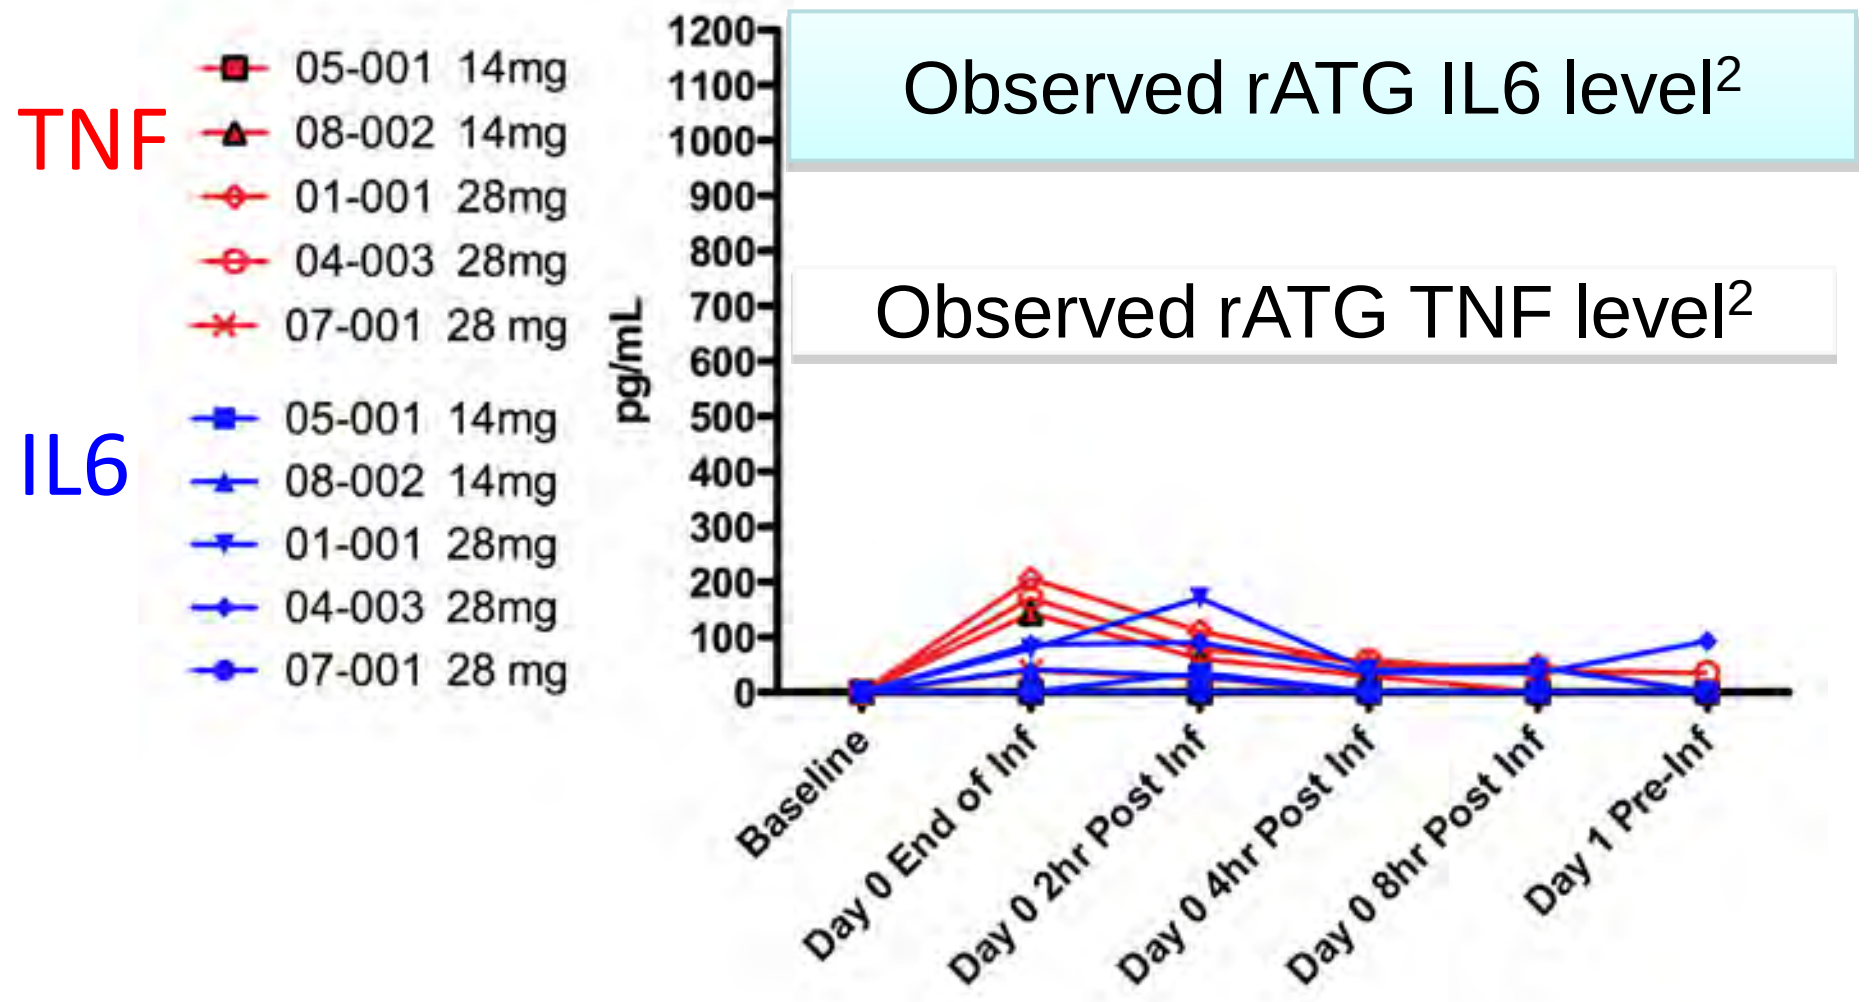

TNF
05-001 14mg
08-002 14mg
01-001 28mg
04-003 28mg
07-001 28 mg
IL6
05-001 14mg
08-002 14mg
01-001 28mg
04-003 28mg
07-001 28 mg
pg/mL
Observed rATG IL6 level2
Observed rATG TNF level2
Baseline
Day 0 End of Inf
Day 0 2hr Post Inf
Day 0 4hr Post Inf
Day 0 8hr Post Inf
Day 1 Pre-Inf

Figure 3. Levels of TNF (A), IL6 (A), IL10, IL1β, IFNγ and IL2 are being determined at multiple time points after infusion. Generally there has been little of any cytokine detected in TOL101 treated subjects. TNF and IL6 have been detected at very low levels, especially when compared to historical rATG data. Furthermore no cytokine release syndrome has been reported. Finally, no human-anti-mouse-antibody has been detected in any TOL101 treated subject.

Conclusions

- **Interim analysis shows that TOL101 is a highly efficient at modulating T cells**
- **TOL101 removes CD3 from the cell surface, with little apparent T cell depeltion**
- **TOL101 inactivated T cells without triggering cytokine release and other SAE's**
- **TOL101 therapeutic profile may aid in overcoming infectious and cancer complications associated with other induction agents**

References

1. Current landscape for T-cell targeting in autoimmunity and transplantation, Getts et al, Immunotherapy, July 2011
2. Pharmacokinetics, Foreign Protein Immune Response, Cytokine Release, and Lymphocyte Subsets in Patients Receiving Thymoglobulin and Immunosuppression,, Guttman, *et al.*, Transplantation Proceedings, 1997

Presented at the 15th Congress of the European Society for Organ Transplantation, 4-7 September 2011, Glasgow, Scotland